Neural Correlates of Gesture Processing Across Development

Elizabeth M. Wakefield & Karin H. James

Department of Psychological and Brain Sciences, Indiana University, Bloomington

Recently, functional Magnetic Resonance Imaging (fMRI) has been utilized to study the processing of speech accompanied by iconic and metaphoric gesture in the adult brain (e.g., Green et al., 2009; Kircher et al., 2009; Straube et al., 2010). Interestingly, the neural mechanisms underlying the processing of iconic and metaphoric gestures differ. What is yet unknown is how the processing of gesture develops. Understanding how children process gesture, as compared to adults, may reveal why children benefit more from learning with gestures than do adults (Hostetter, in press), and increase the overall understanding of how the areas implicated in gesture processing function as an integrated system.

The purpose of the present study is to address the following questions:

(1) Which regions are recruited during iconic and metaphoric gesture processing in the developing brain?

(2) How does this processing change across development, and with increased understanding of sentence content?

Methods

Participants

- 5-6 yr olds (n = 13, *M* = 5.47)
- 7.5-8.5 yr olds (n = 13, *M* = 7.70)
- 10-11 yr olds (n = 13, *M* = 10.50)
- adults (n = 13, *M* = 24.77)

Experimental Session

(1) fMRI simulator exposure (children only)

(2) Scan (anatomical, functional, DTI)

(3) Behavioral session

Stimuli & Functional Run Design

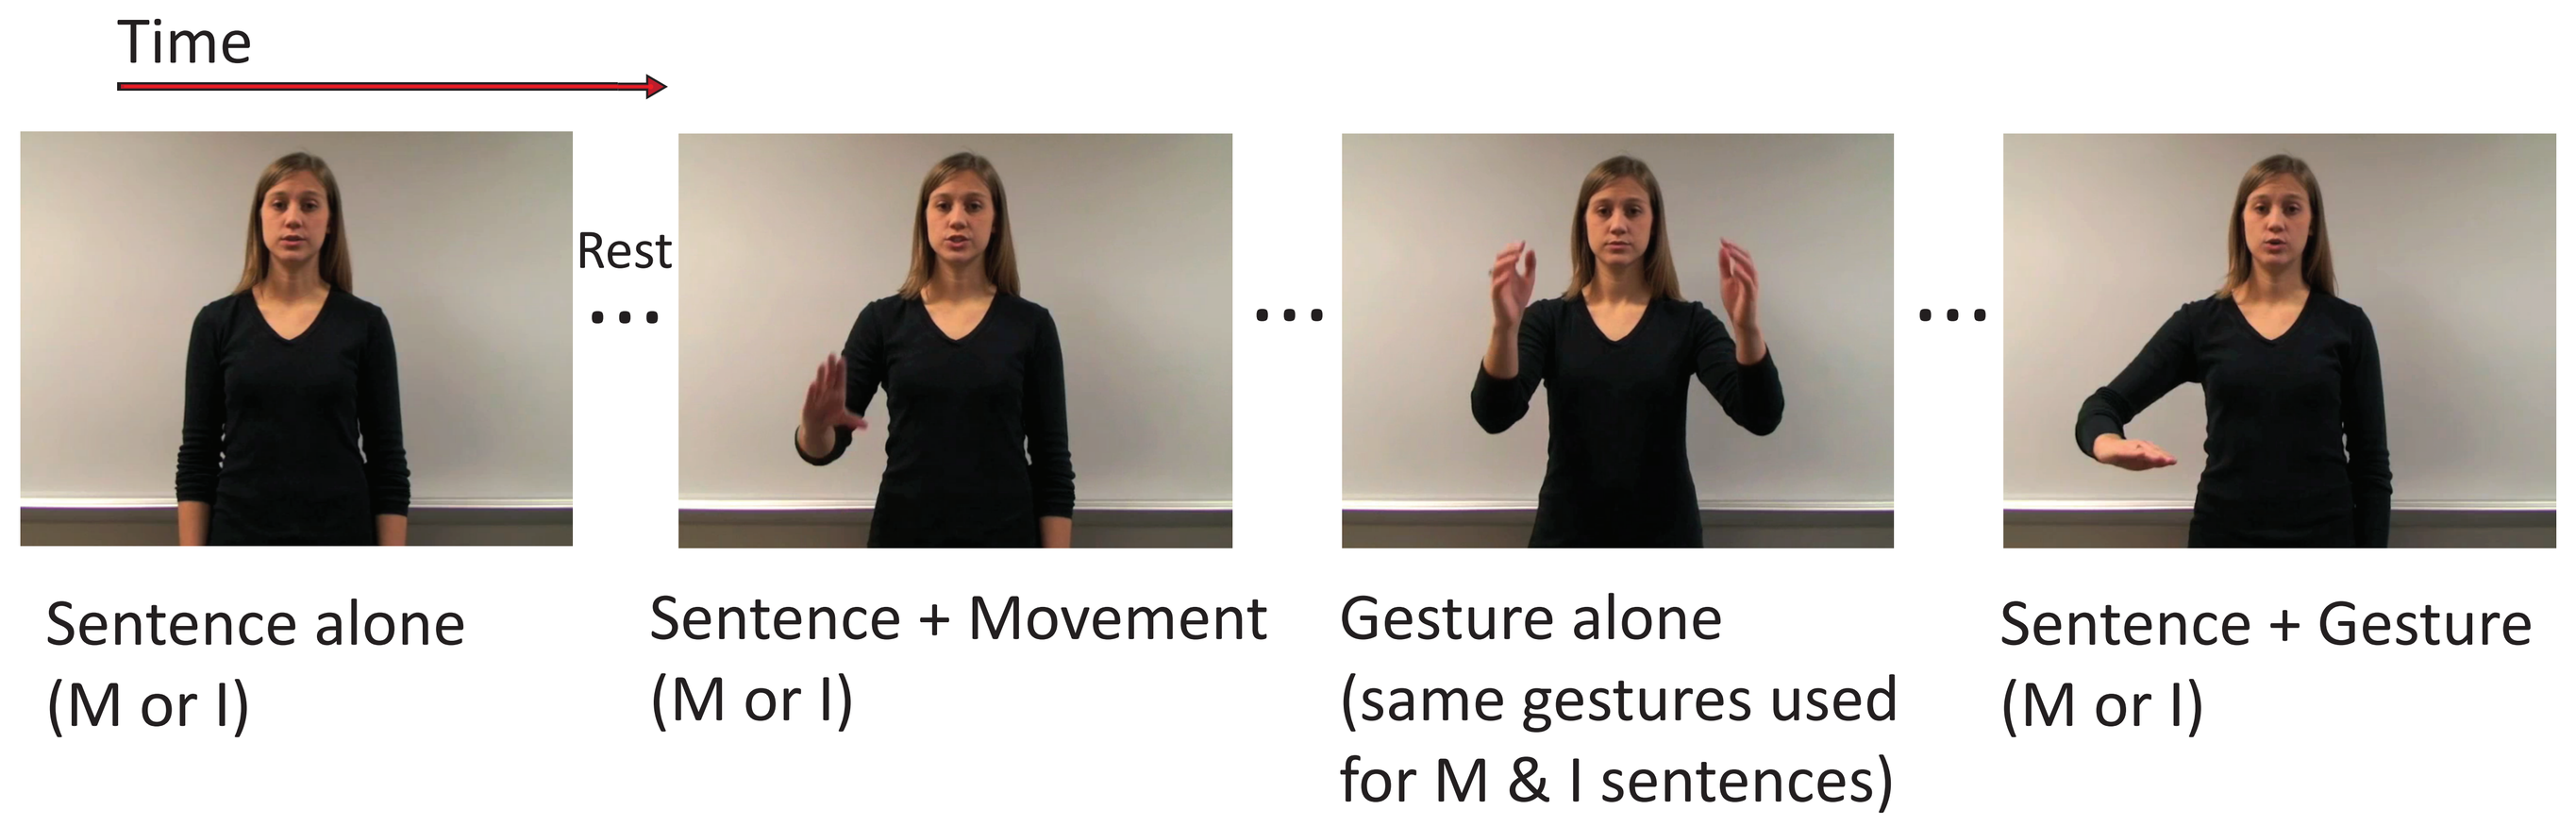
Time
Rest
Sentence alone (M or I)
Sentence + Movement (M or I)
Gesture alone (same gestures used for M & I sentences)
Sentence + Gesture (M or I)

- 2.5 sec videos, 12 per block
- 7 conditions: 3 Metaphoric (M), 3 Iconic (I), 1 M&I

Example Sentences

Iconic
- The teacher ***spoke*** to her.
- The bubble gum ***popped*** onto her face.
- The string was ***wrapped*** around her finger.

Metaphoric
- The book ***spoke*** to her.
- The idea ***popped*** into her head.
- He was ***wrapped*** around her finger.

Results

To identify areas recruited during gesture processing, regardless of speech, a Gesture > Rest contrast was performed.

Adults activated a much larger network than children: Both groups showed middle temporal gyrus (MTG) activation, but adults additionally showed bilateral premotor, superior frontal gyrus (SFG) and superior temporal sulcus (STS) activation, as well as left inferior parietal lobule (IPL) activation.

Two additional whole-brain analyses were conducted to elucidate areas activated for processing gesture in iconic and metaphoric sentence contexts (SG>S, M or I)

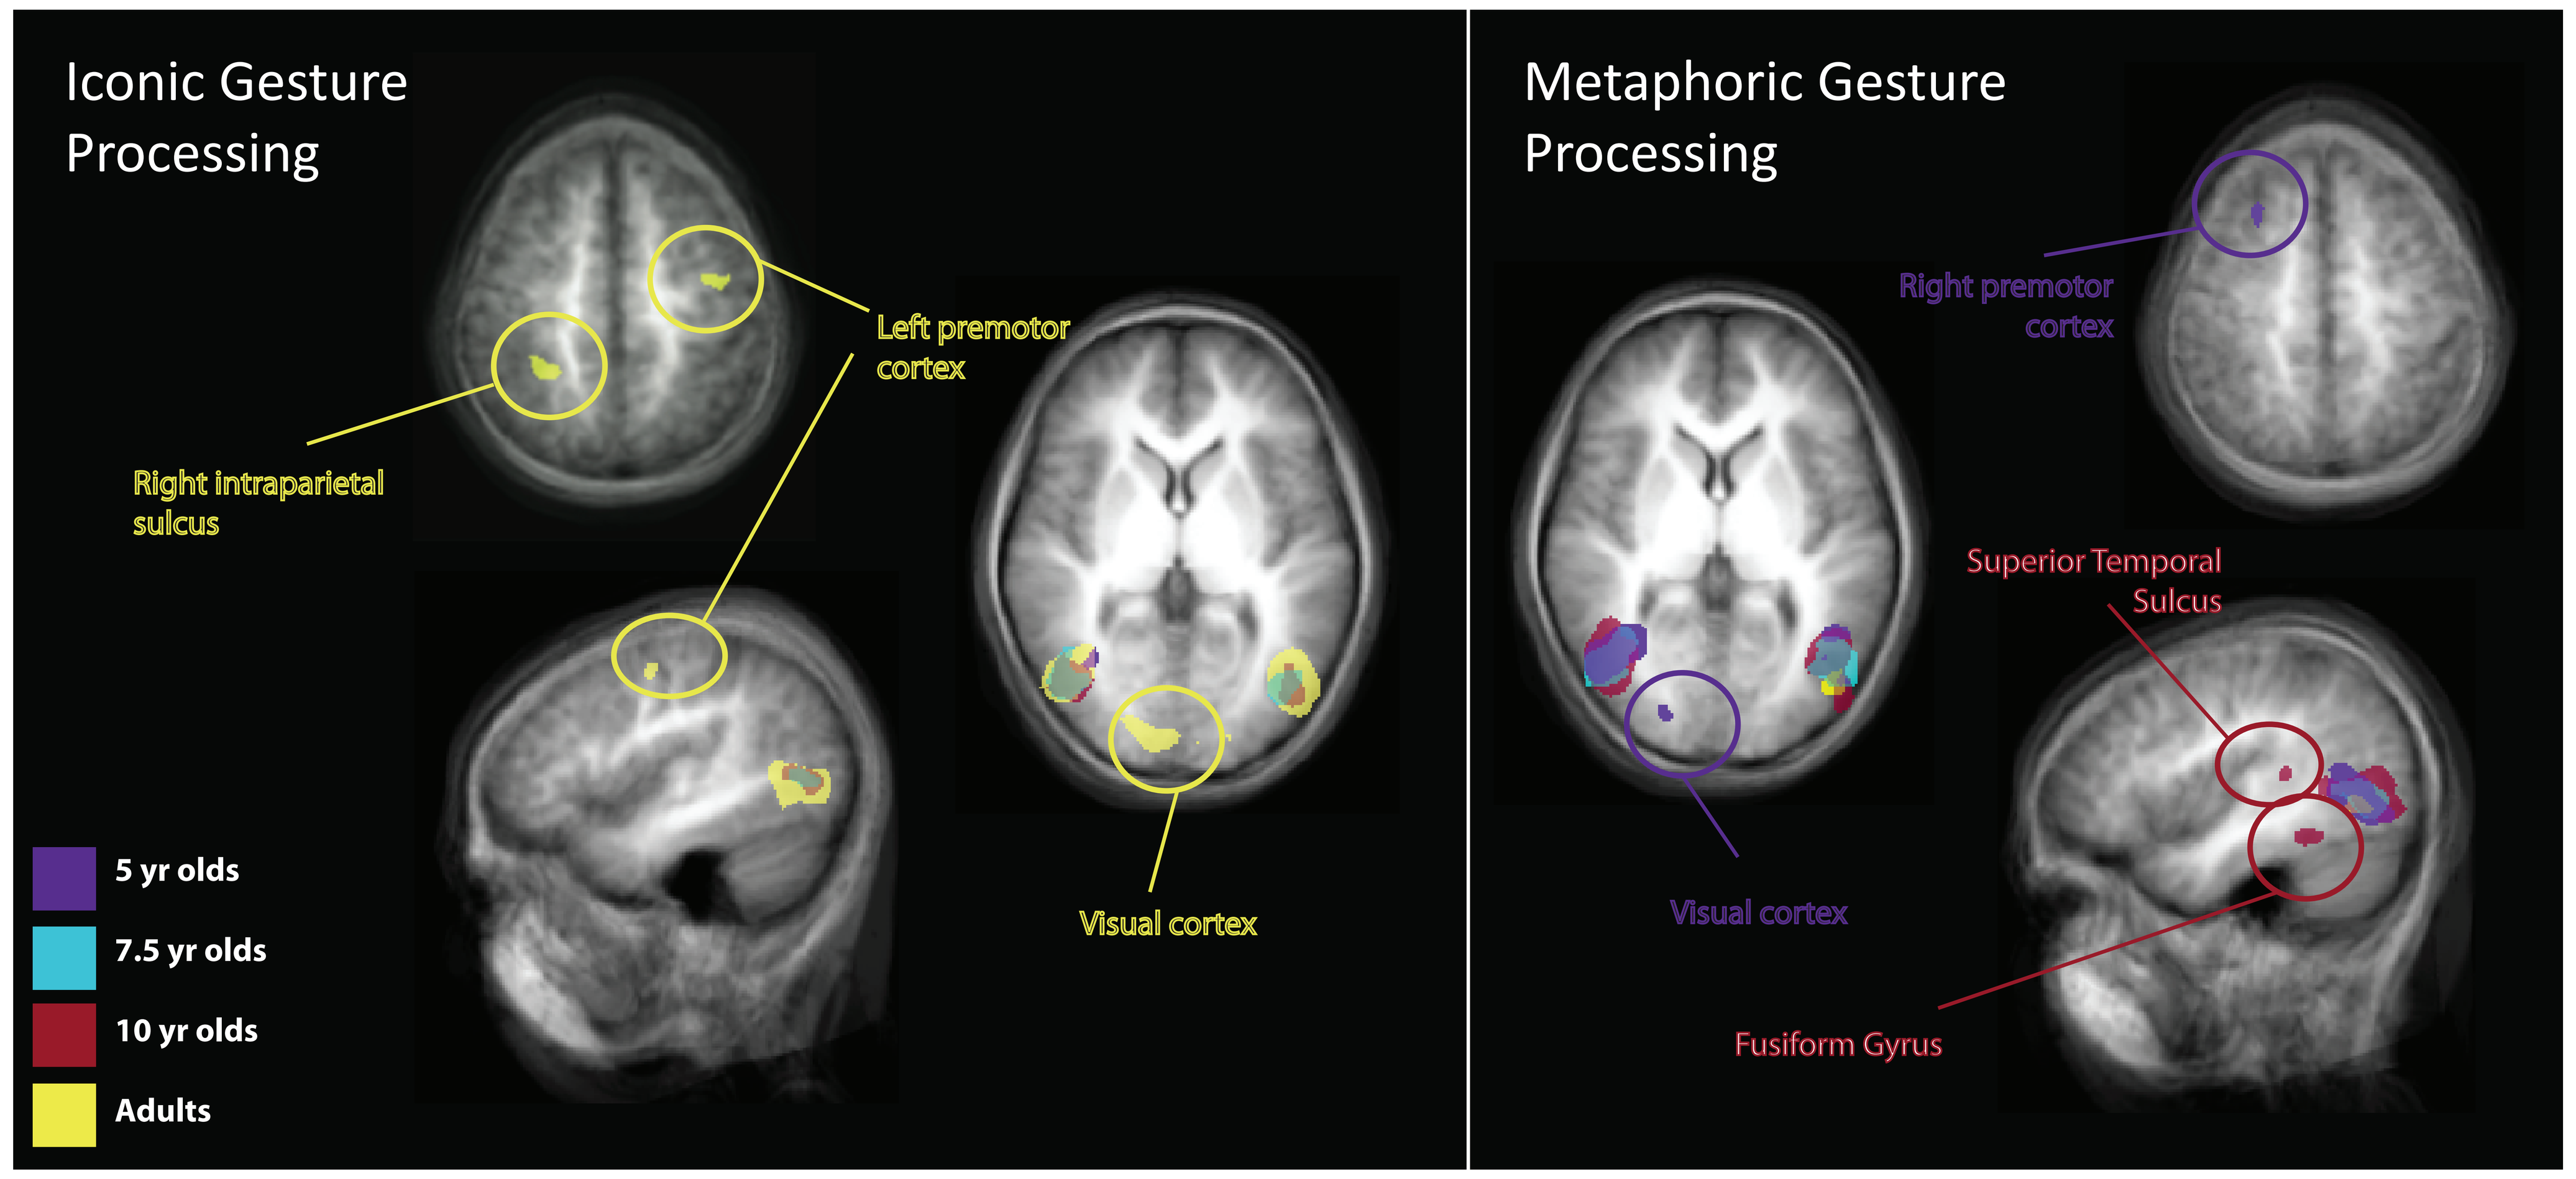
Iconic Gesture Processing
Left premotor cortex
Right intraparietal sulcus
Visual cortex
5 yr olds
7.5 yr olds
10 yr olds
Adults
Metaphoric Gesture Processing
Right premotor cortex
Superior Temporal Sulcus
Visual cortex
Fusiform Gyrus

The only common area of activation for all age groups across all contrasts was the MTG. A ROI analysis was performed for the MTG to determine if this activation differed across conditions for different age groups.

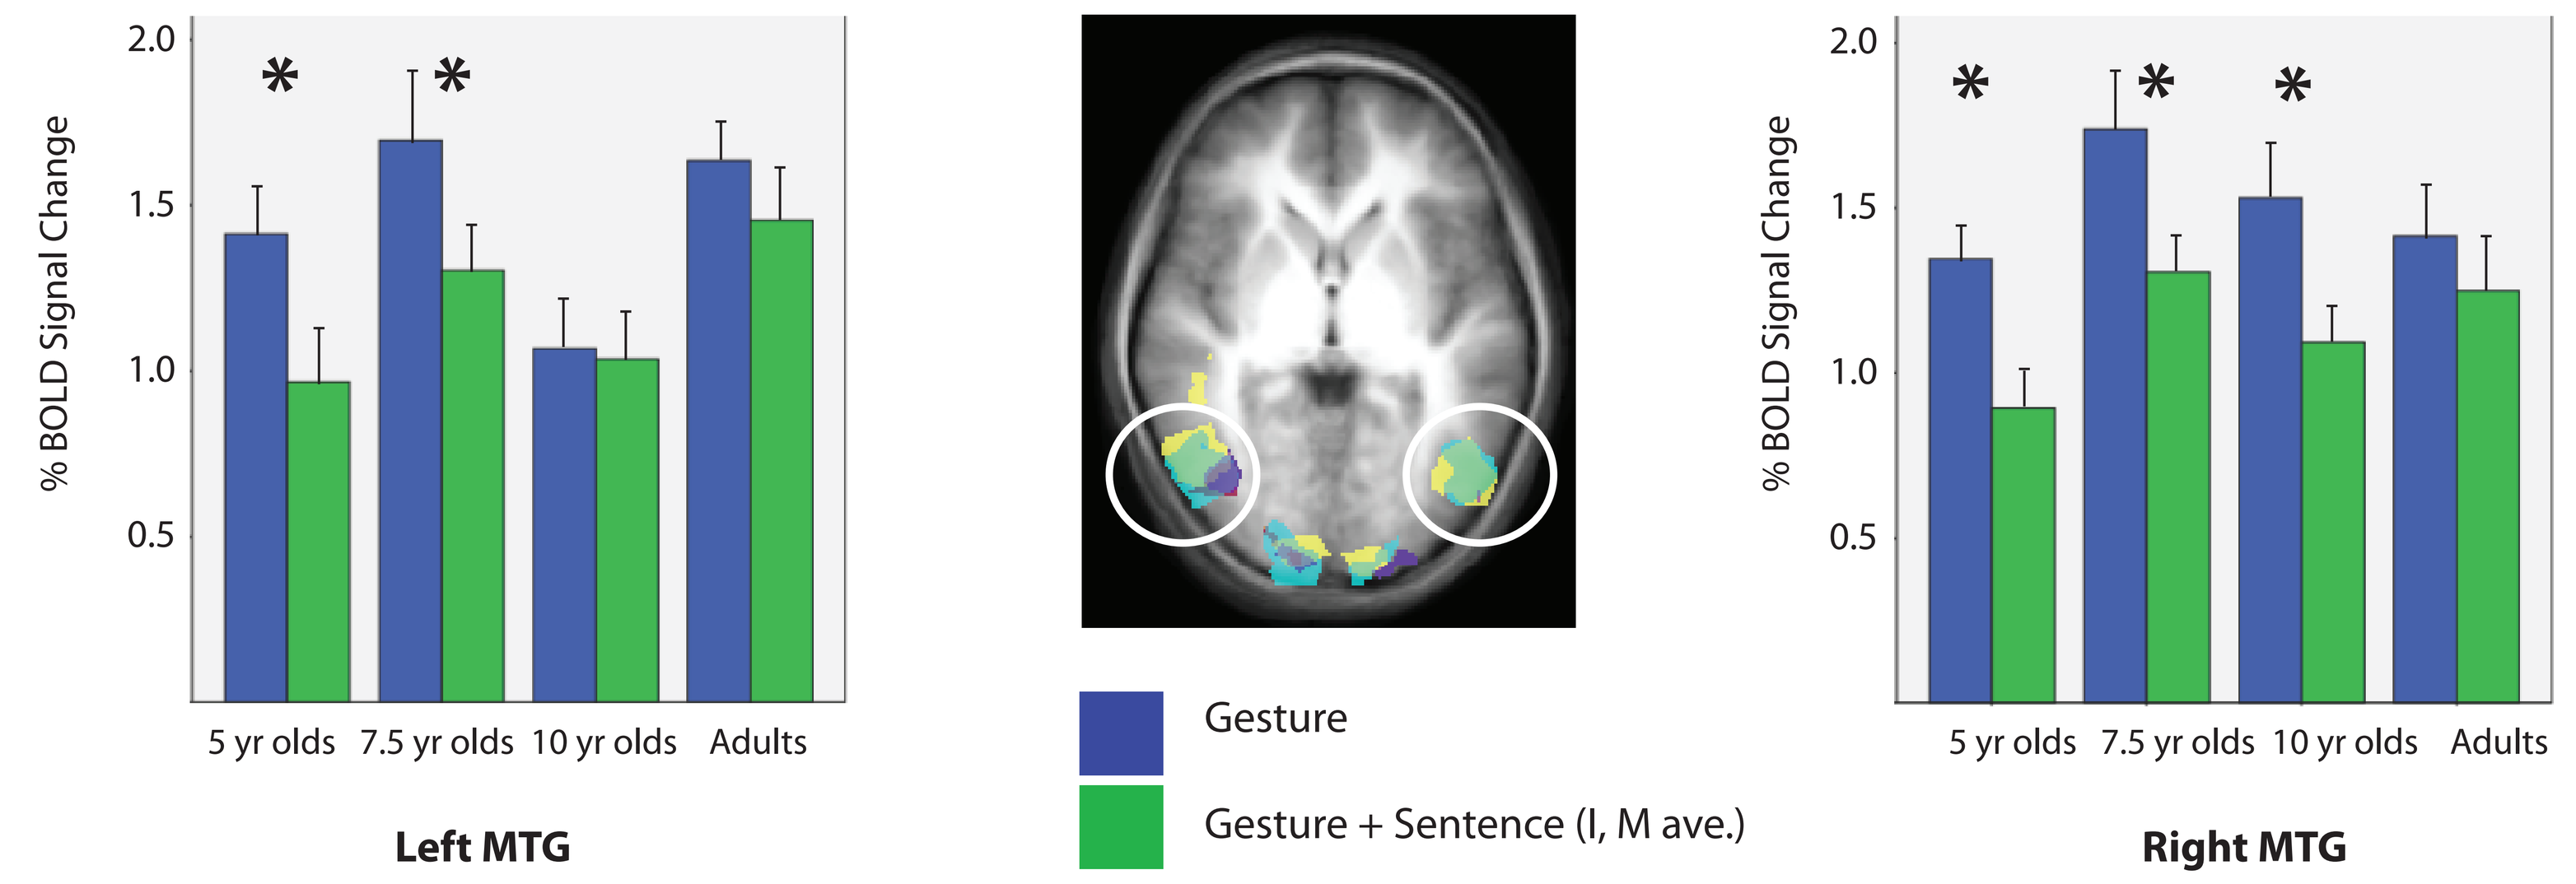
% BOLD Signal Change
2.0
1.5
1.0
0.5
5 yr olds
7.5 yr olds
10 yr olds
Adults
Left MTG
Gesture
Gesture + Sentence (I, M ave.)
Right MTG

- Children younger than 10 show greater MTG activation when watching gesture by itself, than gesture in the context of speech.

- At around 10 years of age, children appear to be transitioning to an adult-like state, in which gesture is processed as easily in the context of a sentence as it is in isolation

Summary

The regions recruited during gesture processing change across development, and in different contexts of gesture use

- When seeing gesture, adults recruit a larger network of regions than children

- This adult network includes motor areas, integration areas, and sensori-motor transformation areas, previously identified as part of this network in other studies

(Dick et al., 2009; Green et al., 2009; Kircher et al., 2009; Skipper et al., 2008; Villarreal et al., 2008; Willems et al., 2007)

- The lack of recruitment of this network by children may indicate a less mature processing system based on less experience with gesture, or less understanding of the meanings to the gestures

- Children DO show recruitment during metaphoric gesture processing--this could be because making sense of the sentences, and their connection to the gestures is more effortful than for adults

Regardless of age or the relation of gesture to speech, the MTG is recruited during gesture processing; however, the degree to which the MTG is recruited in different gesture contexts changes across development

- Children younger than 10 do not show the same amount of MTG activation to gesture when it is in the context of a sentence compared to gesture in isolation

- Perhaps 10 year olds are transitioning to a state at which they can understand abstract relations that younger children cannot, and are beginning to integrate gesture and speech in an adult-like way

References

Dick, A. S., Goldin-Meadow, S., Hasson, U., Skipper, J. I., & Small, S. L. (2009). Co-speech gestures influence neural activity in brain regions associated with processing semantic information. *Human Brain Mapping, 30*, 3509-3526.

Green, A., Straube, B., Weis, S., Jansen, A., Willmes, K., Konrad, K., & Kircher, T. (2009). Neural integration of iconic and unrelated coverbal gestures: A functional MRI study. *Human Brain Mapping, 30*, 3309-3324.

Hostetter, A. B. (in press). When do gestures communicate? *Psychological Bulletin.*

Kircher, T., Straube, B., Leube, D., Weis, S., Sachs, O., Willmes, K., Konrad, K., Green, A. (2009). Neural interaction of speech and gesture: Differential activations of metaphoric co-verbal gestures. *Neuropsychologia, 47*, 169-179.

Skipper, J. I., Goldin-Meadow, S., Nusbaum, H. C., & Small, S. L. (2007). Speech-associated gestures, Broca's area, and the human mirror system. *Brain and Language, 101*, 260-277.

Straube, B., Green, A., Bromberger, B., & Kircher, T. (2010). The differentiation of iconic and metaphoric gestures: Common and unique integration processes. *Human Brain Mapping*

Villarreal, M., Fridman, E. A., Amengual, A., Falasco, G., Gerscovich, E. R., Ulloa, E. R., & Leiguarda, R. C. (2008). The neural substrates of gesture recognition. *Neuropsychologia, 46*, 2371-2382.

Willems, R. M., Özyürek, A., Hagoort, P. (2007). When language meets action: The neural integration of gesture and speech. *Cerebral Cortex, 17*, 2322-2333.

The authors thank our MR technicians, Thea Atwood, Benjamin Pruce, Chris ... Rebecca Ward, and MR physicist Dr. Hu Cheng. We also acknowledge Paroma Bose, Shelley Swain, Alyssa Kersey, Emily Thomas, Caroline Gianesi. This research was supported by the Faculty Research Support Program, awarded to Karin Harman James.

For more information, please contact Elizabeth Wakefield at emwakefi@indiana.edu